our ENZYMES;
NATURE
at WORK

Development of protease deficient *Chrysosporium lucknowense* strains

Vivi Joosten[a], Theo Verwoerd[a], Hans Visser[a], Peter Punt[b] and Jan Wery[a]

[a], Dyadic Netherlands, Nieuwe Kanaal 7-S, 6709 PA Wageningen, The Netherlands. [b], TNO Quality of Life, Utrechtseweg 48, 3704 HE Zeist, The Netherlands.

Introduction

Filamentous fungi such as *Aspergillus niger*, *A. oryzae*, *Trichoderma reesei* and *Chrysosporium lucknowense* (C1) are known to be efficient producers of large quantities of homologous extracellular enzymes. The production levels of heterologous proteins using these fungi is often low as result of the partial or full degradation of the secreted protein by the host proteases. In order to improve the production levels of heterologous proteins, C1 strains are developed that have low protease levels without disturbing their vital functions.

Approach

Previously, C1 was subjected to random mutagenesis and subsequent screening approaches for strain optimization purposes. C1 strains with greatly reduced overall protease activity levels were obtained from both high cellulase production strains as well as newly developed low-cellulase background strains (Fig. 1). These low-cellulase background strains (LC strains) are of special interest because they are able to produce single proteins at high levels.

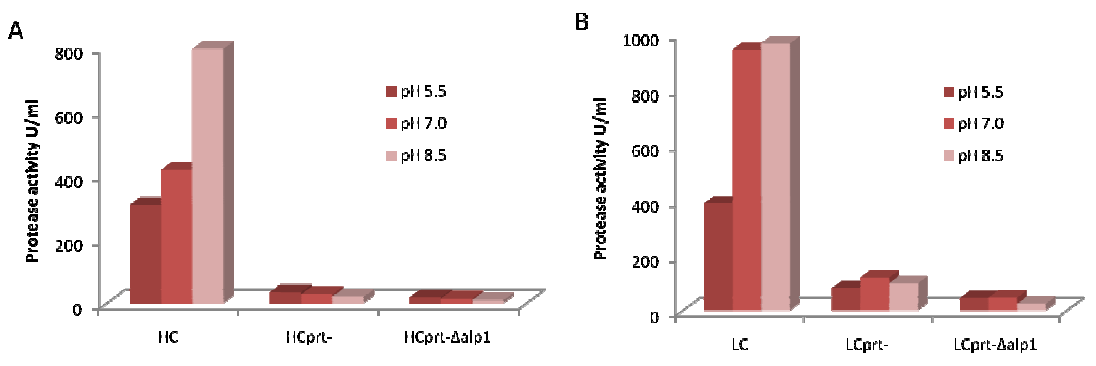
A
B
Protease activity U/ml
pH 5.5
pH 7.0
pH 8.5
HC
HCprt-
HCprt-Δalp1
LC
LCprt-
LCprt-Δalp1

Figure 1. Protease activity in C1 host strains. The activity (U/ml) was measured in end of fermentation samples of the high cellulase production strain HC and its derivatives (A) and LC strains (B). Δ *alp*1 refers to gene disruption of the major secreted protease in *C. lucknowense*.

To further develop the LC strains as host for heterologous protein production, specific proteases in LC culture samples were identified by a combinatorial approach of genome mining, exo-proteome analysis and protease inhibitor studies.

Protease inventory by genome mining and MS analysis

Mining the 38 Mbp C1 genome so far revealed an impressive and diverse set of putative protease encoding genes (Table 1). In the 34 Mbp *Aspergillus niger* genome ~198 genes involved in proteolysis have been identified previously (1) including proteases active in cytosolic, nuclear, proteasomal and lysosomal/vacuolar locations. It is expected that in C1 more putative protease genes of interest are present. Currently the elucidation of the complete proteolytic spectrum is in progress. Additionally, the presence of specific proteases in a LC strain culture sample was shown by ESI-MS/MS analyses. Some of these proteases showed homology to harmful proteases known from other fungal systems such as *A. niger* PEPA, PEPB and PEPE (2).

Table 1. Classification of putative C1 protease genes. The genes are classified into peptidase families, according to the MEROPS peptidase database.

Peptidase family	# genes	MS analysis	Examples *of A. niger* homologues (2)
Aspartic (*e.g. pep*4 and *pep*7)	17	1	PEPA; PEPE
Cysteine	2	1	
Glutamic (e.g.*prt*1)	4	0	PEPB
Metallo	13	12	PEPH
Serine (*e.g. alp*1 and *prt*8)	22	9	PEPC; PEPD; PEPF

Protease inhibitor studies

Incubations of LC fermentation culture filtrates with protease-sensitive proteins and protease inhibitors were used to gain further insight in the type of peptidases that were present (Fig. 2). Proteolysis of a heterologous protein X, for example, was prevented in the presence of antipain and chymostatin, which are inhibitors of cysteine and serine type proteases.

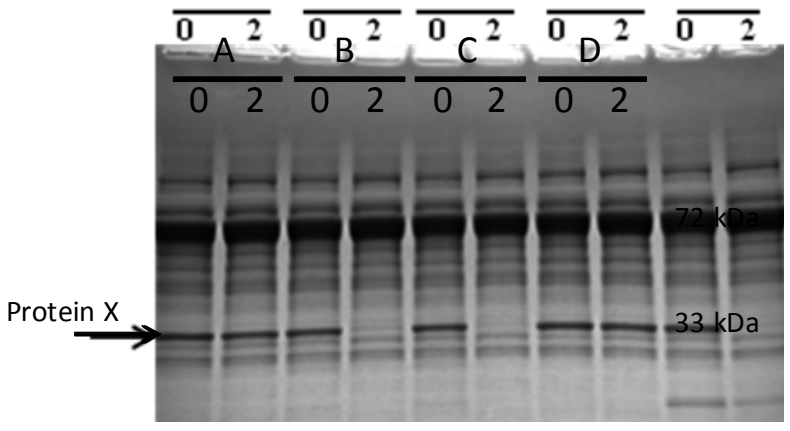
A
B
C
D
0 2 0 2 0 2 0 2
72 kDa
33 kDa
Protein X

Figure 2. SDS-PAGE analysis of protein X stability in the presence of protease inhibitors. A) antipain, B) pepstatin A, C) bestatin D) chymostatin. Time samples were taken at t = 0 and t = 2 hrs. The position of the target protein X is indicated with an arrow.

Targeted protease gene disruptions

Several C1 strains have been developed that are low in overall protease activity by random mutagenesis and screening procedures. Based on the current protease inventory, several protease encoding genes were selected for gene disruption in order to further reduce the protease activity. Using specially designed Δku70 strains that promote targeted integration of knock-out cassettes, several target protease genes have been deleted (Fig. 3).

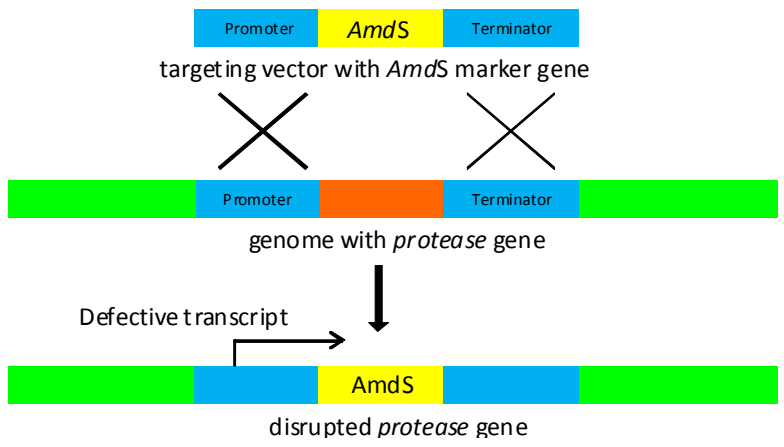
Promoter
*Amd*S
Terminator
targeting vector with *Amd*S marker gene
Promoter
Terminator
genome with *protease* gene
Defective transcript
AmdS
disrupted *protease* gene

Figure 3. Targeted protease disruption strategy by homologous recombination. The *amd*S selection marker was flanked by homologous protease DNA sequence and used in LC transformation experiments. This work was greatly facilitated by the use of strains defective in the non-homologous-end-joining DNA pathway (NHEJ). The frequency of obtaining the correct gene disruptions was significantly increased in Δku70 strains (30 - 40%) when compared to WT strains (1 - 2 %).

Conclusions

Several *C. lucknowense* host strains containing one or more protease gene disruptions have been generated. This has paved the way towards efficient production of heterologous proteins.

Contact: **Dr. V. Joosten (vjoosten@dyadic.nl)**
Dr. J. Wery (jwery@dyadic.nl)

References
1) Pel *et al.* (2007). *Nat Biotechnol.* 25(2):221-231
2) van den Hombergh *et al.* (1997). *Trends Biotechnol.* 15(7):256-263

www.dyadic.nl